Arginase 1 Deficiency Masquerading as Hereditary Spastic Paraplegia: Implications for Diagnostic Testing

Poster number: **510**

UT Southwestern Medical Center

MC McNutt,[1] MW Bechter,[2] AM Trucillo[2]

[1]UT Southwestern Medical Center, Dallas, TX, USA; [2]Aeglea BioTherapeutics, Inc., Austin, TX, USA

Background

- Arginase 1 deficiency (ARG1-D) is a genetic disorder caused by mutations in the *ARG1* gene and subsequent deficiency of the arginase 1 enzyme that catalyzes hydrolysis of arginine in the last step of the urea cycle[1,2] (**Figure 1**)
- ARG1-D causes progressive spastic diplegia, which typically manifests in early childhood. Other manifestations include developmental delay, intellectual disability, and seizures[3-5] (**Figure 2**)

Figure 1. Metabolic Impact of Arginase 1 Deficiency

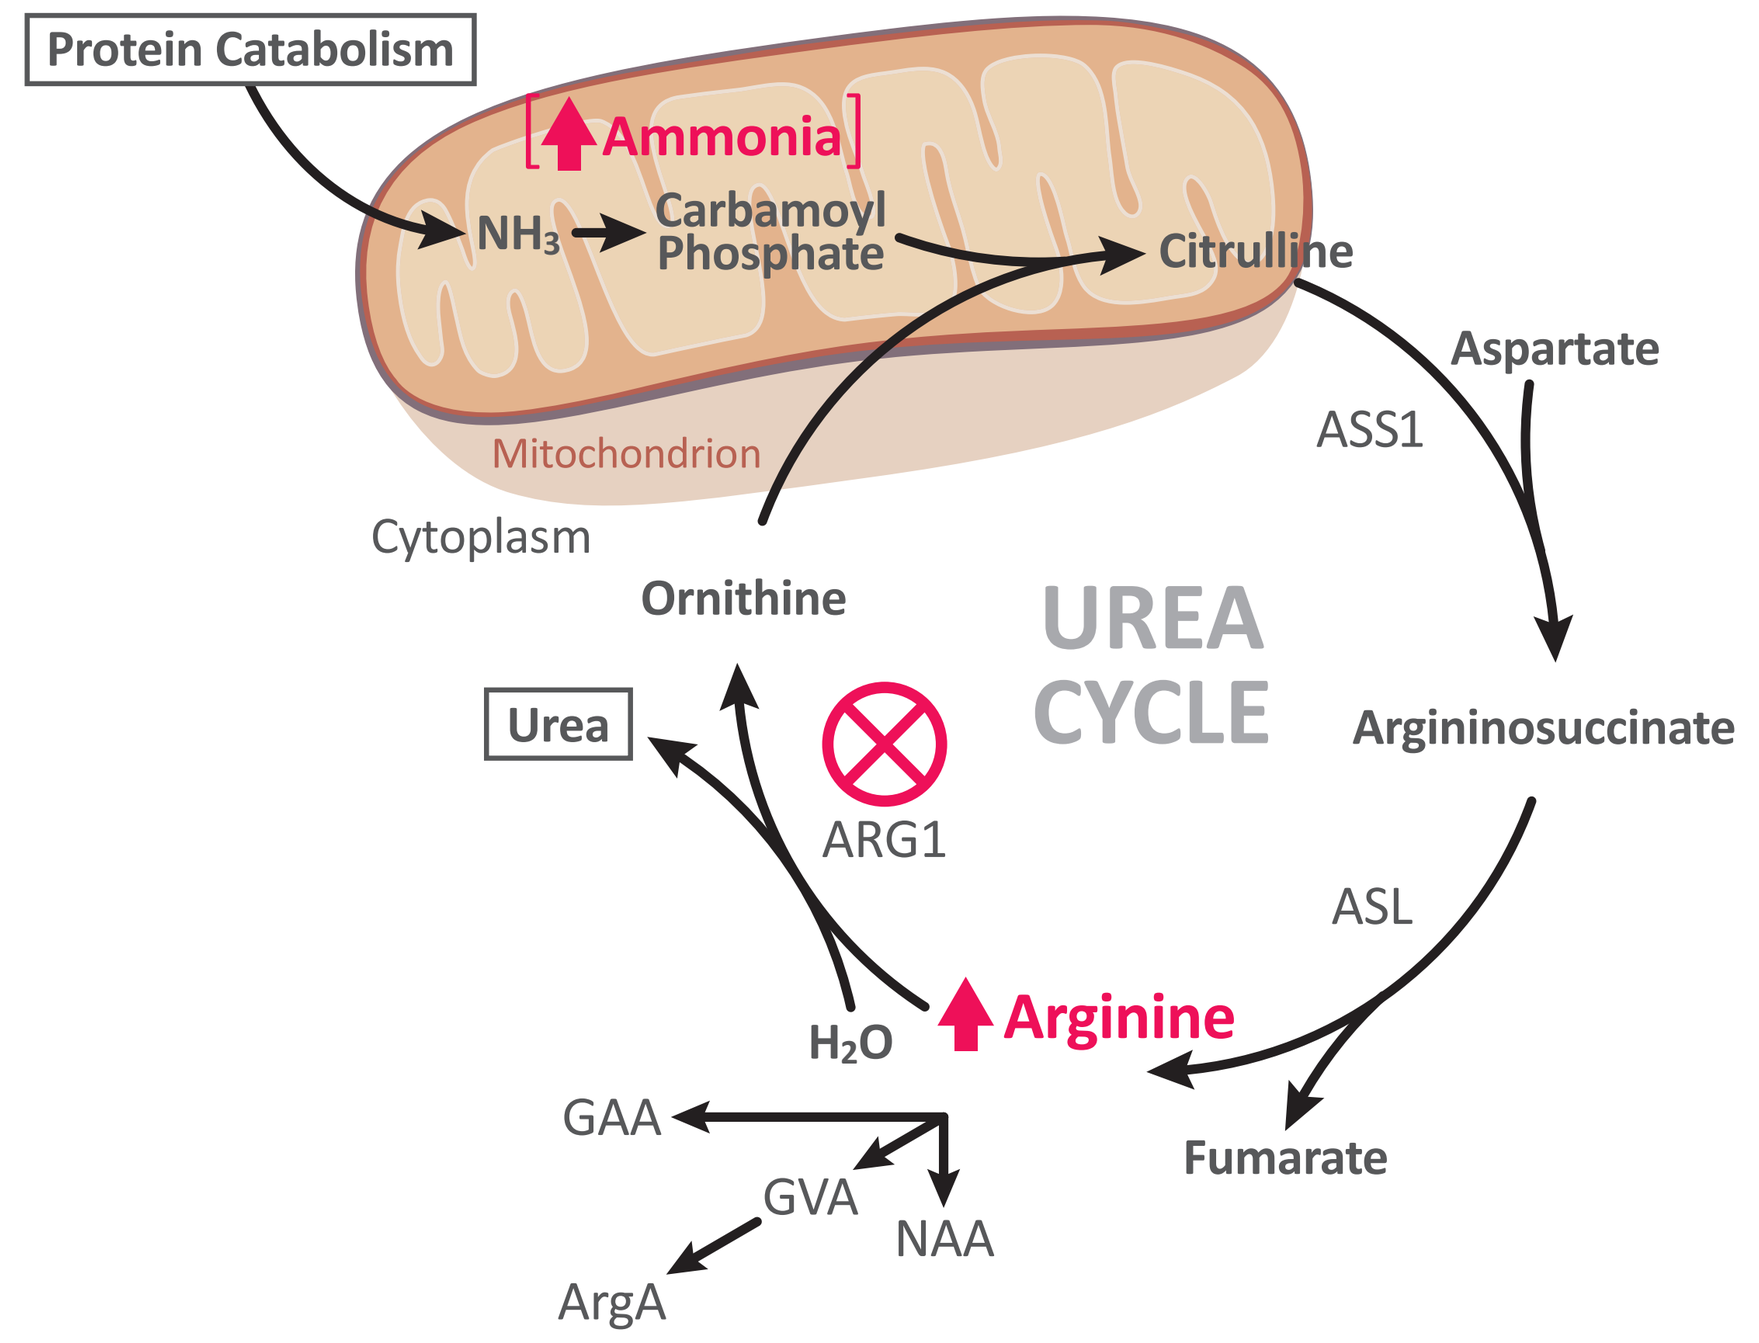
Protein Catabolism
Ammonia
NH_3
Carbamoyl Phosphate
Citrulline
Aspartate
ASS1
Mitochondrion
Cytoplasm
Ornithine
UREA CYCLE
Argininosuccinate
Urea
ARG1
ASL
H_2O
Arginine
GAA
GVA
NAA
Fumarate
ArgA

ARG1, arginase 1; ASL, argininosuccinate lyase; ASS1, argininosuccinate synthase 1. Arginine-derived metabolites: ArgA, argininic acid; GAA, guanidinoacetic acid; GVA, α-keto-δ-guanidinovaleric acid; NAA, N-acetyl-L-arginine.

Figure 2. Typical Clinical Onset of ARG1-D

Birth to 1 year[2,3,5]	Infancy (2–5 years)[1-5]	Childhood (5–10 years)[1-5]
• Initial 6–12 months often uneventful • May present with seizures, episodes of mild hyperammonemia: irritability, feeding difficulties, poor appetite, nausea/vomiting, decreased alertness	• Spasticity in lower limbs (tip-toe gait) • Seizures • Cognitive impairment (delay/regression) • Spontaneous avoidance of high-protein foods	• Progressive spasticity • Variable decline in growth • Variable decline in neuromotor and cognitive skills – Loss of gait – Decrease/loss of vocabulary/language – Loss of sphincter control

- Increased plasma arginine is the hallmark of ARG1-D; arginine levels may be 3.5- to 10-fold higher than those in unaffected individuals.[6] If ARG1-D is suspected, diagnosis can be confirmed via plasma arginine levels, through genetic testing, or through enzymatic assays for red blood cell arginase activity
- Delays in diagnosis or misdiagnosis often occur because of lack of disease awareness and differences in patient presentation
 - Although hyperammonemia occurs in many urea cycle disorders, severe hyperammonemia is less frequently seen in ARG1-D,[5] and manifestations of ARG1-D are driven by persistently elevated arginine
 - ARG1-D may be misdiagnosed as hereditary spastic paraplegia (HSP) because both disorders are characterized by progressive lower-extremity spastic paraparesis[3] (**Table 1**)
 - ARG1-D is not generally considered in the differential diagnosis of HSP and *ARG1* may not be included in HSP gene-sequencing panels

Table 1. Differential Characteristics of ARG1-D vs HSP

Symptom	ARG1-D	HSP
Normal arginine, ammonia, and urea	✘	✔
Avoidance of high-protein food	✔	✘
Seizures	✔	✘
Urinary urgency	✘	✔

ARG1-D, arginine 1 deficiency; HSP, hereditary spastic paraplegia.

- We report a patient who was undiagnosed or misdiagnosed as having HSP for >3 years before being diagnosed with ARG1-D

Case Study

Clinical Presentation

- A 24-year-old woman presented with a 4-year history of slowly progressive leg weakness and recurrent falls (**Figure 3**)
- The patient was referred to neurology, where magnetic resonance imaging (MRI) of the spine was normal, but MRI of the brain revealed nonspecific, fluid-attenuated inversion recovery hyperintensities in the bilateral frontal periventricular white matter and mild cortical atrophy (**Figure 4**)

Figure 3. Patient Timeline

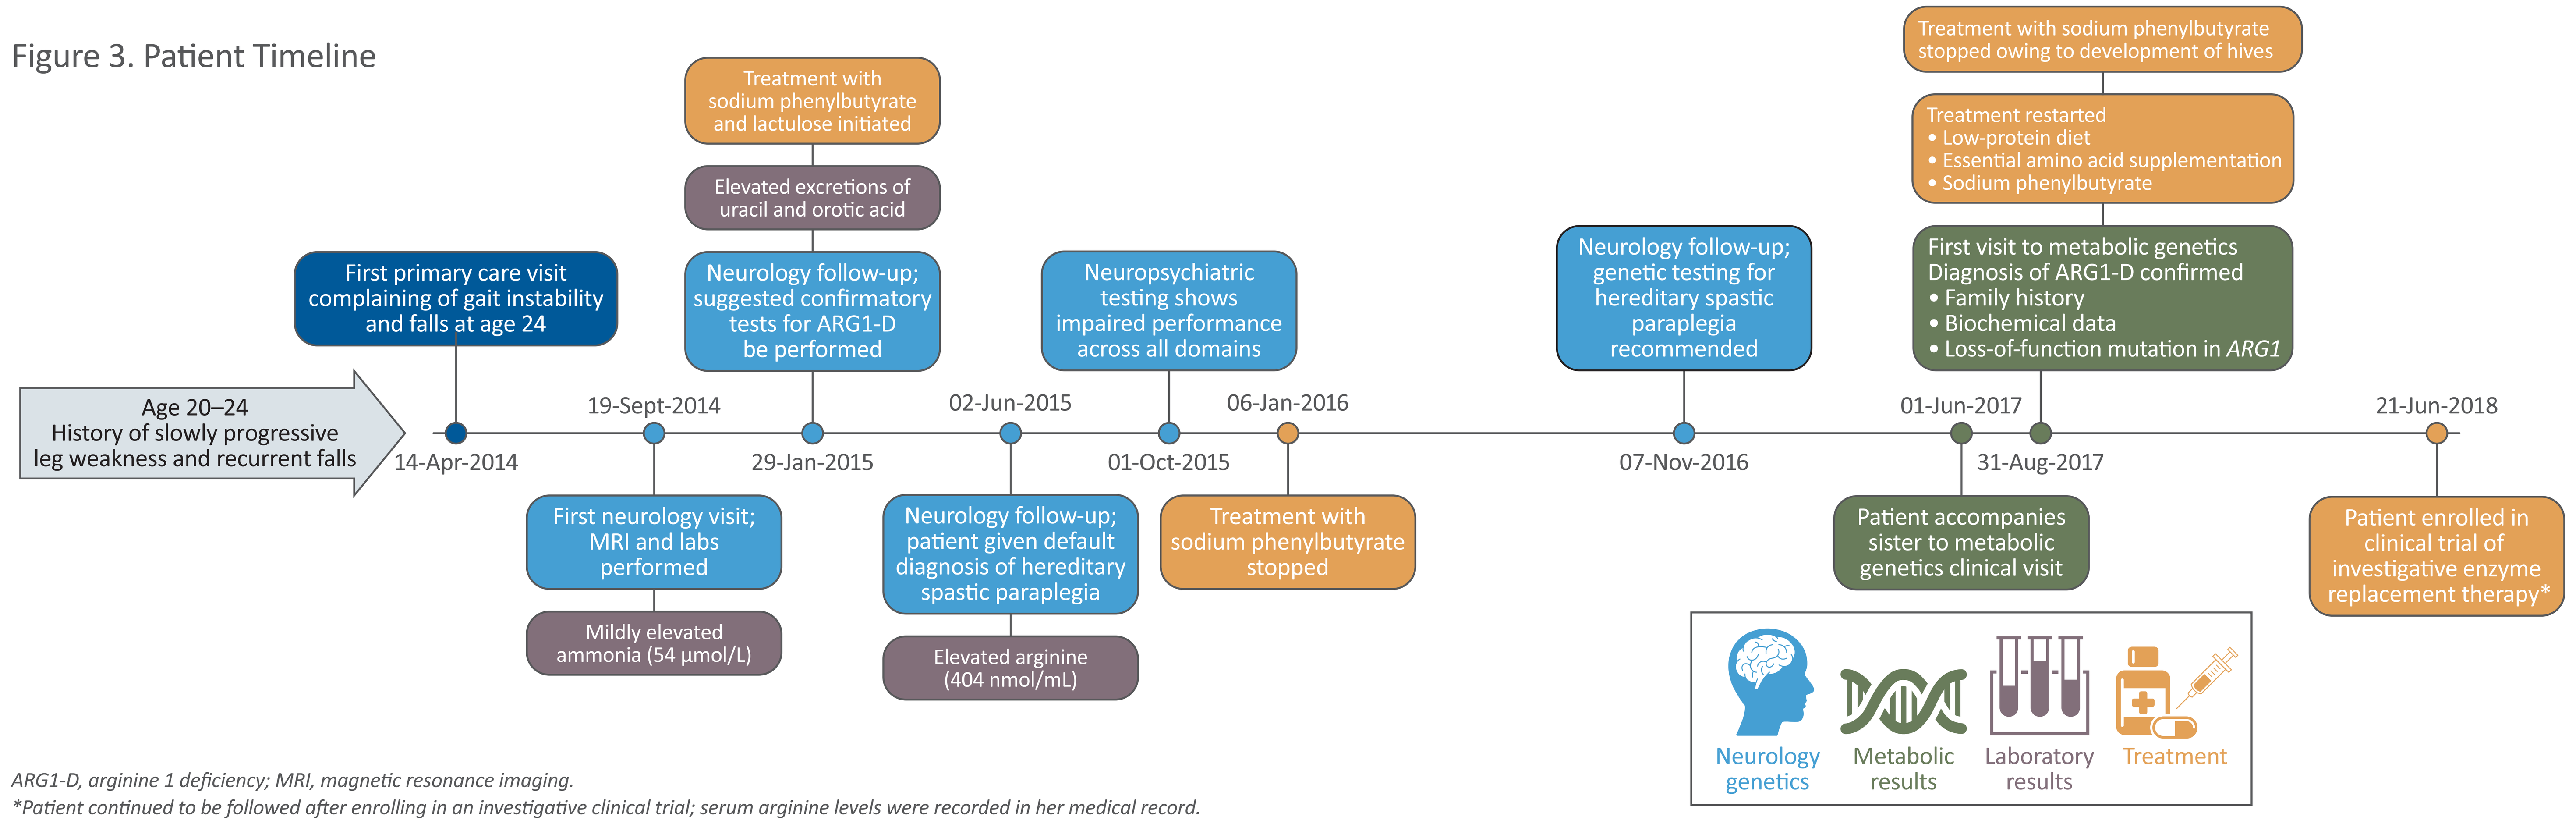
Age 20–24
History of slowly progressive leg weakness and recurrent falls
14-Apr-2014
First primary care visit complaining of gait instability and falls at age 24
19-Sept-2014
First neurology visit; MRI and labs performed
Mildly elevated ammonia (54 μmol/L)
29-Jan-2015
Neurology follow-up; suggested confirmatory tests for ARG1-D be performed
Elevated excretions of uracil and orotic acid
Treatment with sodium phenylbutyrate and lactulose initiated
02-Jun-2015
Neurology follow-up; patient given default diagnosis of hereditary spastic paraplegia
Elevated arginine (404 nmol/mL)
01-Oct-2015
Neuropsychiatric testing shows impaired performance across all domains
06-Jan-2016
Treatment with sodium phenylbutyrate stopped
07-Nov-2016
Neurology follow-up; genetic testing for hereditary spastic paraplegia recommended
01-Jun-2017
Patient accompanies sister to metabolic genetics clinical visit
31-Aug-2017
First visit to metabolic genetics
Diagnosis of ARG1-D confirmed
• Family history
• Biochemical data
• Loss-of-function mutation in ARG1
Treatment restarted
• Low-protein diet
• Essential amino acid supplementation
• Sodium phenylbutyrate
Treatment with sodium phenylbutyrate stopped owing to development of hives
21-Jun-2018
Patient enrolled in clinical trial of investigative enzyme replacement therapy*
Neurology genetics
Metabolic results
Laboratory results
Treatment

ARG1-D, arginine 1 deficiency; MRI, magnetic resonance imaging.
**Patient continued to be followed after enrolling in an investigative clinical trial; serum arginine levels were recorded in her medical record.*

Figure 4. MRI of the Brain Showing (A) Mild Cortical Atrophy and (B) Nonspecific T2-FLAIR Hyperintensities

A B

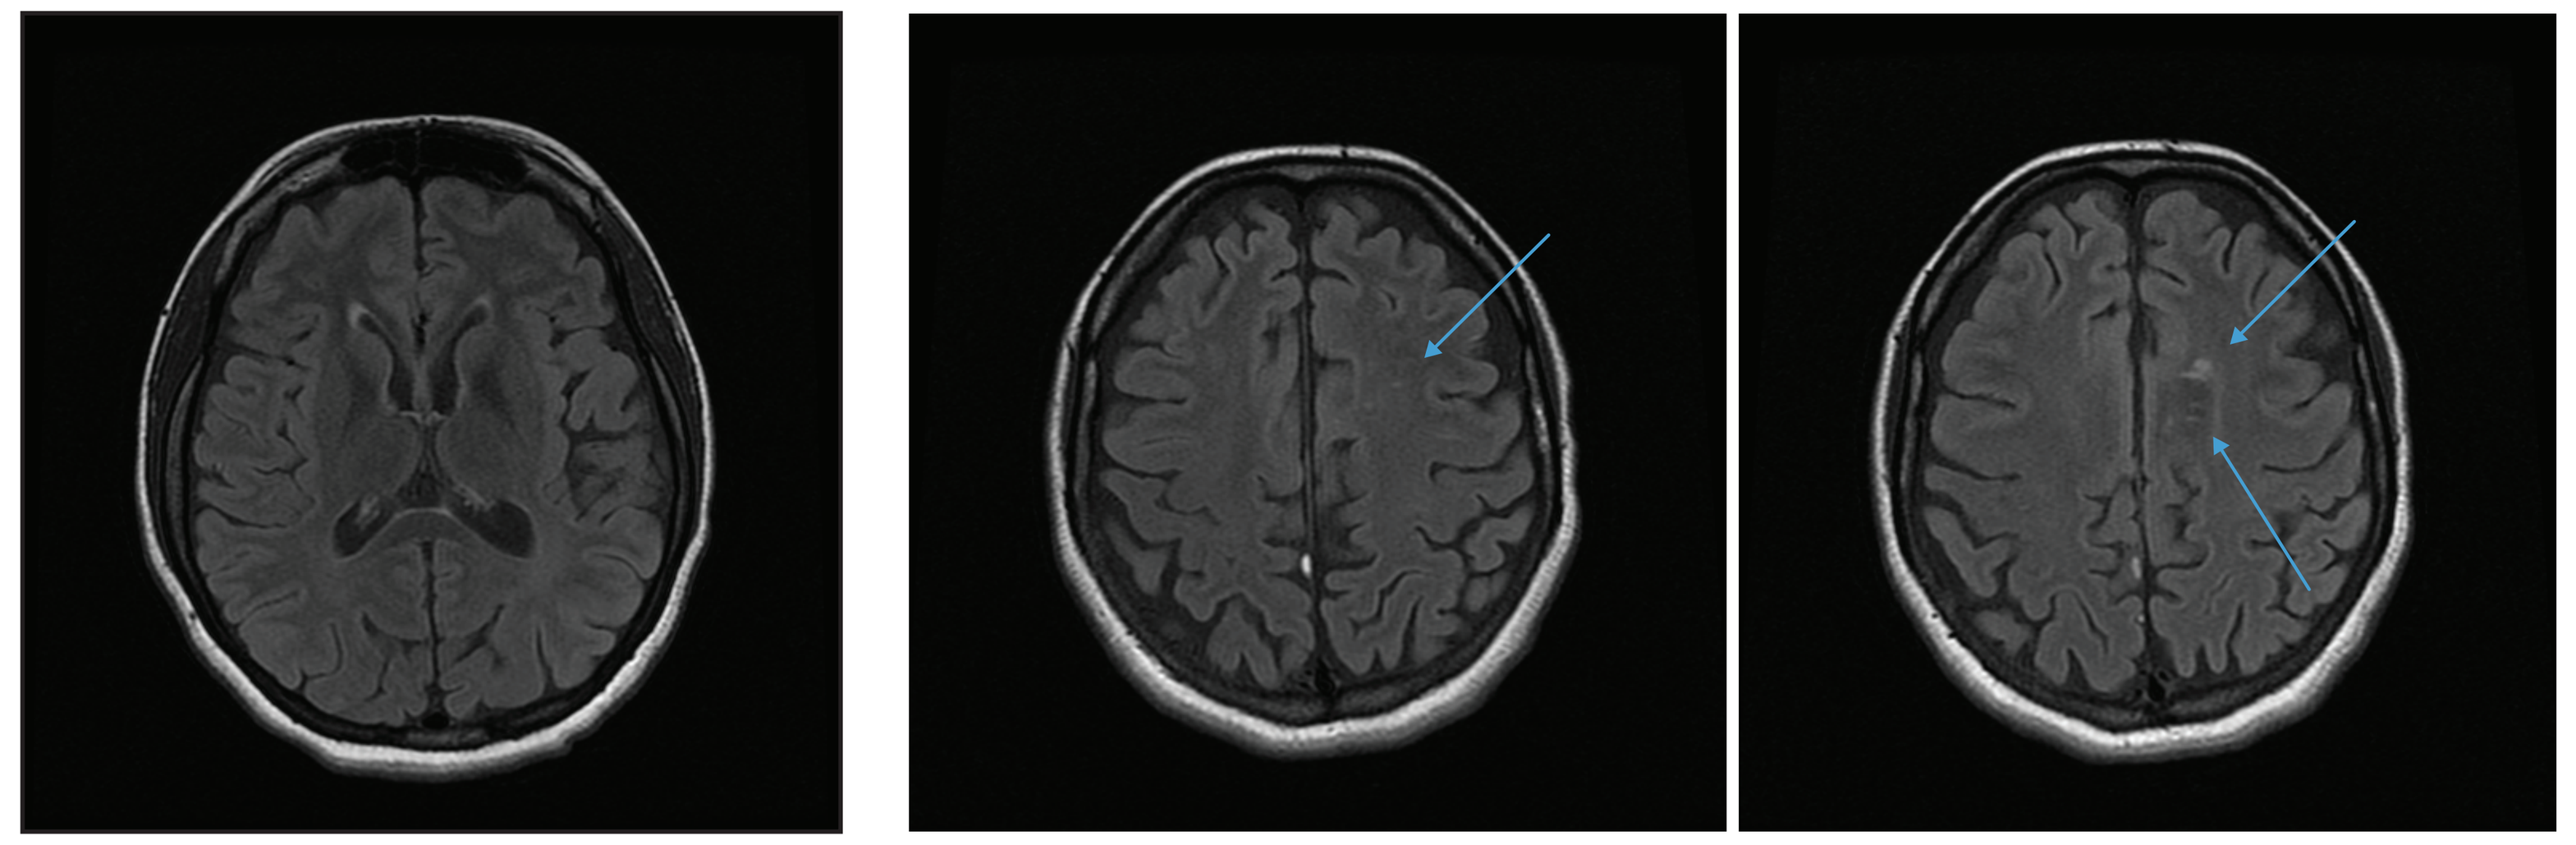

T2-FLAIR fluid-attenuated inversion recovery; MRI, magnetic resonance imaging.

- In biochemical evaluations
 - Only mild elevation of ammonia (54 μmol/L) was observed
 - Biotin, vitamin B12, copper, and vitamin E levels were normal
 - Very long chain fatty acids were normal
 - Urine organic acids identified increased uracil and orotic acid excretion
- Because of her family history (an older sister with a diagnosis of ARG1-D based on biochemical testing), ARG1-D was considered highest on the diagnostic differential and treatment with the nitrogen scavenger sodium phenylbutyrate and lactulose was initiated
- At a neurology follow-up, it was noted that the patient should be evaluated for biotinidase deficiency, vitamin E deficiency, or arginase deficiency; the default diagnosis was HSP
 - Testing revealed elevated plasma arginine (404 nmol/mL; **Table 2**)

Table 2. Serum Amino Acids Outside of Reference Range*

Organic Acid	Value (reference), nmol/mL
Aminobutyric acid	8 (9–37)
Alanine	145 (200–579)
Arginine	404 (32–120)
Asparagine	30 (37–92)
Glutamic acid	12 (13–113)
Homocitrulline	3 (<2)
Isoleucine	26 (36–107)
Leucine	44 (68–183)
Lysine	42 (103–255)
Ornithine	17 (38–130)
Phenylalanine	23 (35–80)
Proline	65 (63–187)
Serine	57 (63–187)
Taurine	28 (42–156)
Threonine	32 (85–231)
Tryptophan	27 (29–77)
Tyrosine	23 (31–90)
Valine	82 (136-309)

**Reflects levels at the time of the second neurology follow-up visit.*

- At a subsequent neurology follow-up, the patient had continued to decline, with progression of lower-extremity spasticity, and underwent additional evaluation for potential causes of these symptoms. At this time, treatment with the nitrogen scavenger and lactulose was stopped owing to lack of clinical benefit
- Despite the patient's family history, HSP remained the suspected diagnosis for >2 years, with no discussion of a disorder of arginine metabolism as the primary diagnosis in the patient's medical record

Diagnosis of ARG1-D

- At age 27, the patient accompanied her sister to a visit with a genetics provider and was subsequently diagnosed with ARG1-D during a metabolic genetics clinical visit based on biochemical data, family history, and homozygous loss-of-function mutations (rs1064794165) in the *ARG1* gene
- Following diagnosis of ARG1-D, the patient was placed on a regimen of severe protein restriction and treated with essential amino acid supplementation and a nitrogen scavenger (sodium phenylbutyrate). Attempts to lower plasma arginine levels were not sufficiently effective (**Table 3**)

Table 3. Serum Arginine Levels*

Date	Arginine level, nmol/mL (reference: 32–120)
02-Jun-2015	404
01-Sept-2017	422
01-Nov-2017	463
30-Nov-2017	418
22-Feb-2018	497

**Data reflect information in the patient's medical record.*

- The patient is currently enrolled in a clinical trial of an investigative therapy, pegzilarginase, a human enzyme–based approach to lower plasma arginine levels

Conclusions

- Previous reports have described misdiagnosis of ARG1-D with spastic diplegia as HSP, largely owing to similarities in neurological presentation[3,7]
- Consistent with the literature, the patient described in this case report was misdiagnosed for multiple years despite her family history of ARG1-D, allowing establishment and progression of typical manifestations of the disorder
- Because ARG1-D (unlike HSP) is a treatable cause of spastic paraplegia, there is a continued need for increased awareness of the disease among healthcare providers and improved recognition of when biochemical and/or genetic testing should be performed

Informed Consent The patient provided informed consent and data were collected under a protocol (STU-2018-0103) approved by the Institutional Review Board of UT Southwestern Medical Center.

Acknowledgments The authors thank RM Rao for his contributions to this work. Editorial support was provided by Catherine DeBrosse, PhD, of Curry Rockefeller Group, LLC (Tarrytown, NY), and was funded by Aeglea BioTherapeutics, Inc. (Austin, TX).

Funding This study was funded by Aeglea BioTherapeutics, Inc. (Austin, TX).

Disclosures **MC McNutt** has received funds for clinical studies, honoraria, and travel reimbursement from Aeglea Biotherapeutics, Inc. **MW Bechter** and **AM Trucillo** are Aeglea employees.

References **1.** Crombez EA and Cederbaum SD. *Mol Genet Metab.* 2005;84(3):243-251. **2.** Sin YY, et al. *J Mol Med (Berl).* 2015;93(12):1287-1296. **3.** Carvalho DR, et al. *Pediatr Neurol.* 2012;46(6):369-374. **4.** Cai X, et al. *Medicine (Baltimore).* 2018;97(7):e9880. **5.** Scaglia F and Lee B. *Am J Med Genet C Semin Med Genet.* 2006;142c(2):113-120. **6.** Diez-Fernandez C, et al. *Hum Mutat.* 2018;39(8):1029-1050. **7.** Lee BH, et al. *Pediatr Neurol.* 2011;44(3):218-220.

JOINT 16TH INTERNATIONAL CHILD NEUROLOGY CONGRESS AND 49TH ANNUAL CHILD NEUROLOGY SOCIETY VIRTUAL MEETING • OCTOBER 12–23, 2020